CM-101 DEMONSTRATES REDUCTION IN SERUM FIBROTIC BIOMARKERS IN A PHASE 1b RANDOMIZED, CONTROLLED MULTIPLE DOSE TRIAL IN NAFLD PATIENTS

Rifaat Safadi[1], Adi Mor[2], Michal Segal-Salto[2], Neta Barashi[2], Yafit Sadot[2], Lina Krasny[2], Sharon Hashmueli[2], Arnon Aharon[2]

(1) Liver unit, Hadassah medical center, Israel. (2) ChemomAb Ltd.

AASLD November 13-16, 2020
The Liver Meeting®
Digital Experience

Background

CCL24 is a chemokine that was shown to be involved in the development of liver fibrosis. Blocking CCL24 attenuated liver fibroblast activation and significantly reduced liver fibrosis in multiple PSC and NASH animal models. This activity supports CCL24's potential role as a therapeutic target for liver fibrotic diseases.

CM-101 is a fully humanized CCL24 blocking monoclonal antibody that showed a favorable safety profile in a single administration study in healthy volunteers. Here we report data from a phase 1b repeated administration study in NAFLD patients demonstrating CM-101's safety profile and effect on fibrotic bio-markers.

Study Design

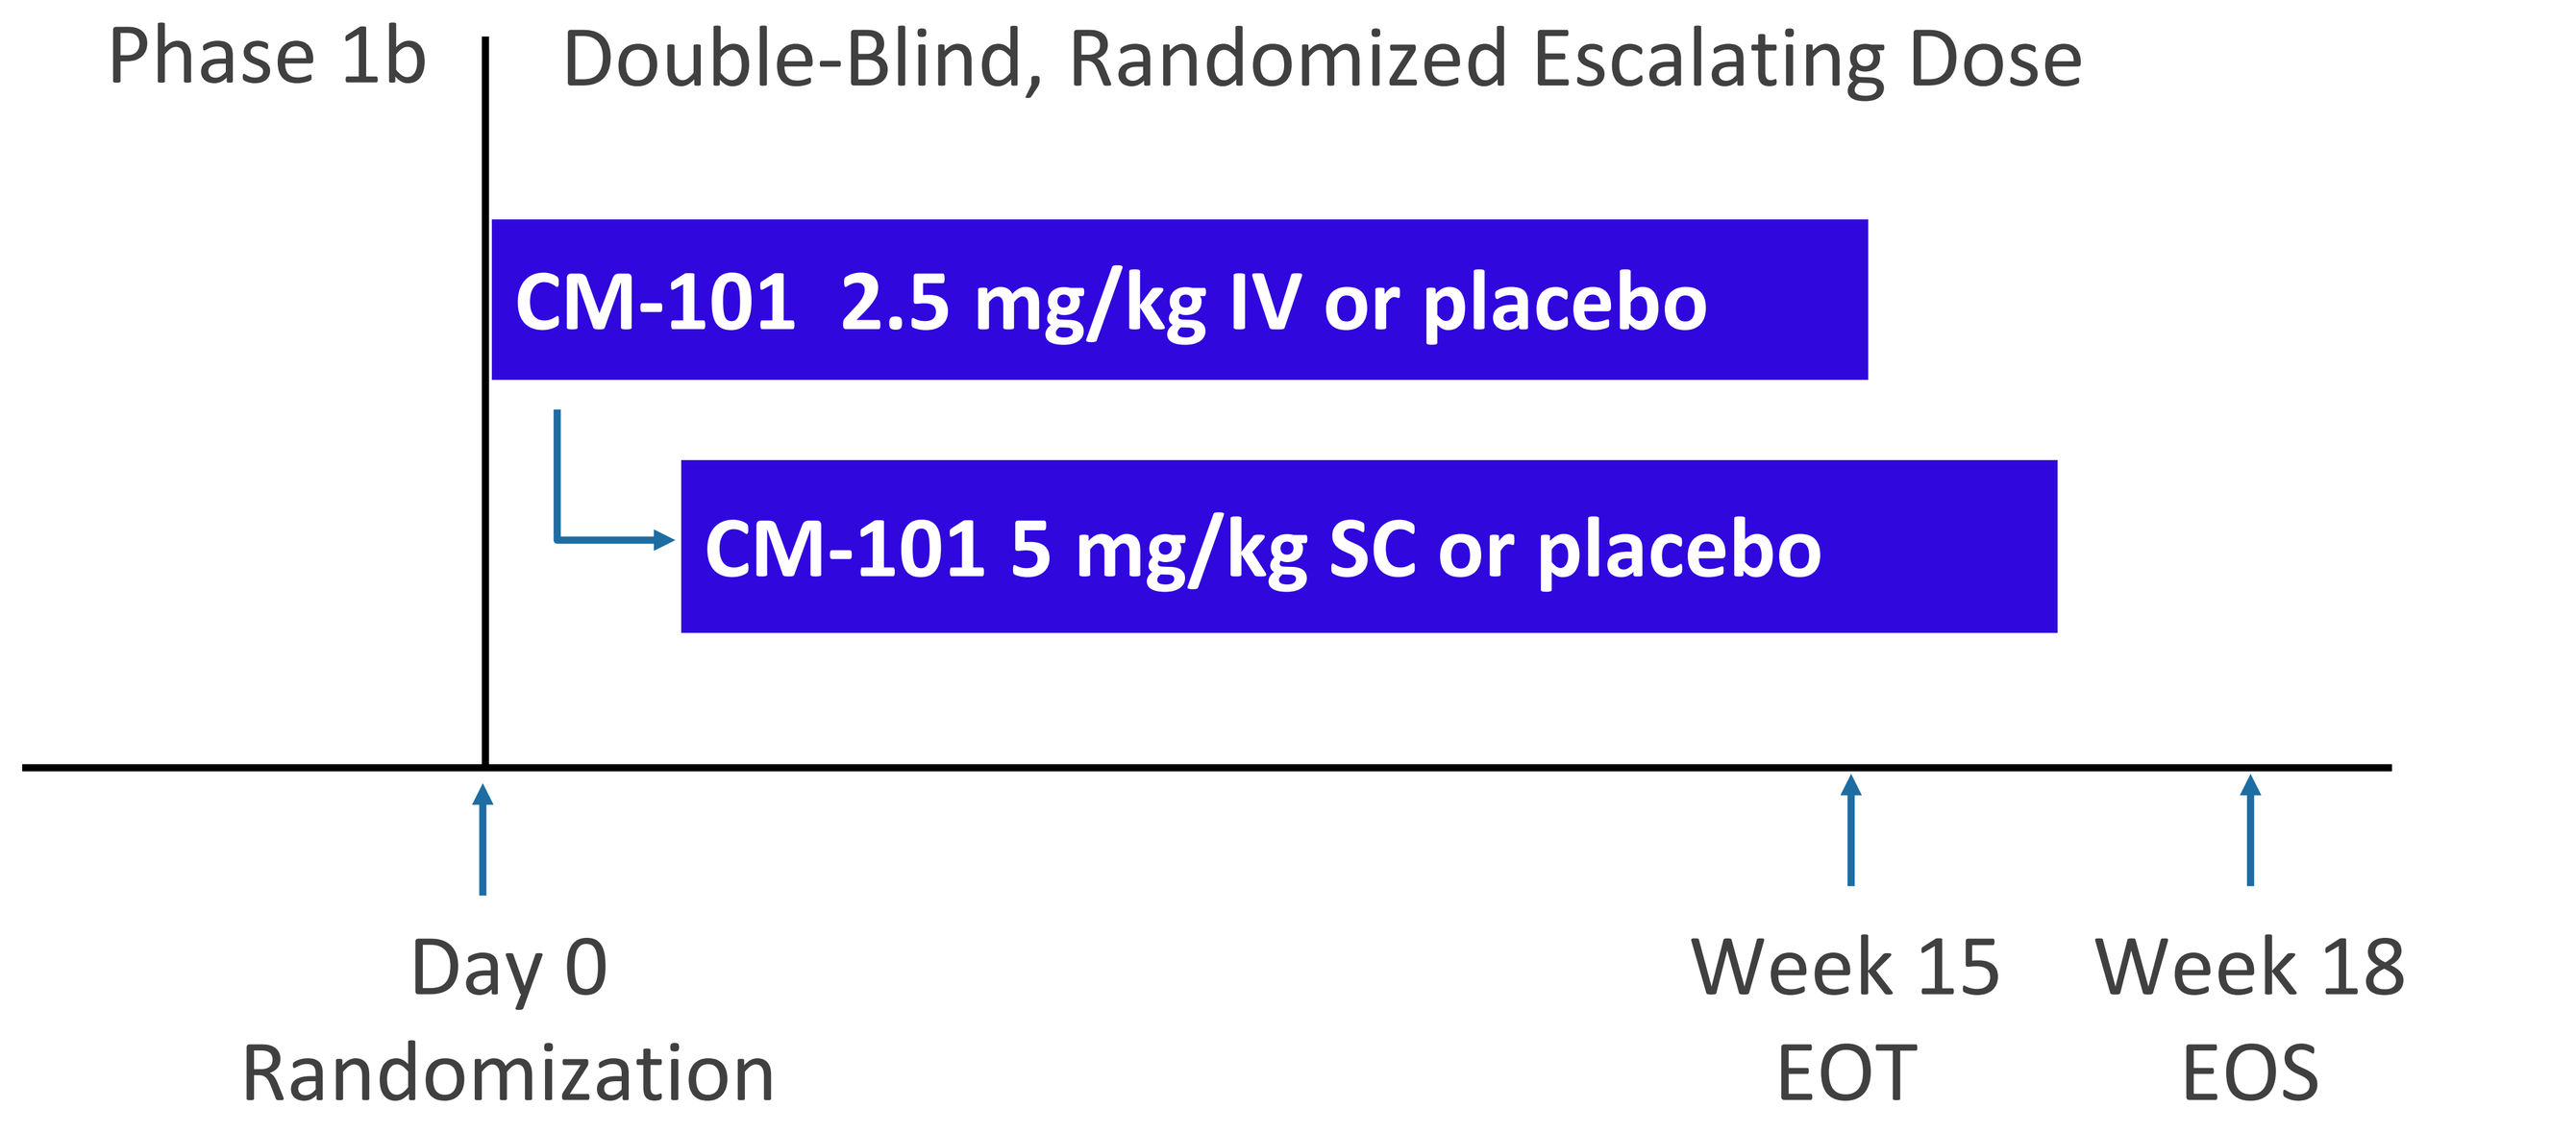
Phase 1b
Double-Blind, Randomized Escalating Dose
CM-101 2.5 mg/kg IV or placebo
CM-101 5 mg/kg SC or placebo
Day 0
Randomization
Week 15
EOT
Week 18
EOS

Methods

This controlled randomized single-center Phase 1b study assessed safety, tolerability, PK, anti-drug antibodies and exploratory pharmacodynamics of multiple CM-101 administrations in 16 NAFLD patients with normal liver enzymes. Patients received five CM-101 treatments (every 3 weeks), either as IV 2.5 mg/kg (6 *vs.* 2 matching placebo cases) or SC 5 mg/kg (6 *vs.* 2) and had a post treatment follow-up period of 42 days.

Serum samples were analyzed for:

- Collagen turnover markers (Pro-C3, Pro-C4, C3M) at Nordic biosciences, Denmark.
- Fibrotic bio-markers (TIMP1, TIMP2, PDGF AA), using multiplex (Millipore) at Chemomab Ltd. R&D lab, Israel.
- Pharmacokinetic (PK) was measured by ELISA for CM-101 serum levels at KCAS, US.

Elastography was preformed using Fibroscan™ at the Hadassah clinical center liver unit.

Demographics

	CM-101 (N=12)	Placebo (N=4)	All (N=16)
Age (± SD)	47.0 ± 10.7	57.2 ± 10.1	49.5 ± 10.8
Male (n)	50% (6)	75% (3)	56%
Caucasian	100%	100%	100%

Safety

In this study 5 repeated administrations of CM-101 were found to be safe and well tolerated for both tested doses and administration modes. All reported AEs were mild or moderate (no sever AE's were reported). One non-drug related SAE reported in the SC dose group (meningioma) that led to early patient discontinuations. No injection site reactions were reported for both SC and IV administrations.

Conclusion

In this phase 1b repeated administration study, 12 weeks of CM-101 treatment was safe and well tolerated. Dose proportional increase in CM-101 exposure for both 2.5 mg/kg IV and 5 mg/kg SC was demonstrated accompanied by target engagement.

We present, for the first time in human, signals of anti-fibrotic activity following CM-101 treatment. The reduction of serum biomarkers and improvement of elastography, in low disease burden NAFLD patients, is encouraging and supports the anticipated anti fibrotic activity mediated by CCL24 blockage.

CM-101 is currently tested in a phase 2a study in PSC and a bio-marker proof of mechanism study in NASH is planned.

Analysis of non-invasive fibrotic bio-markers and PK - PD

Collagen turnover markers*

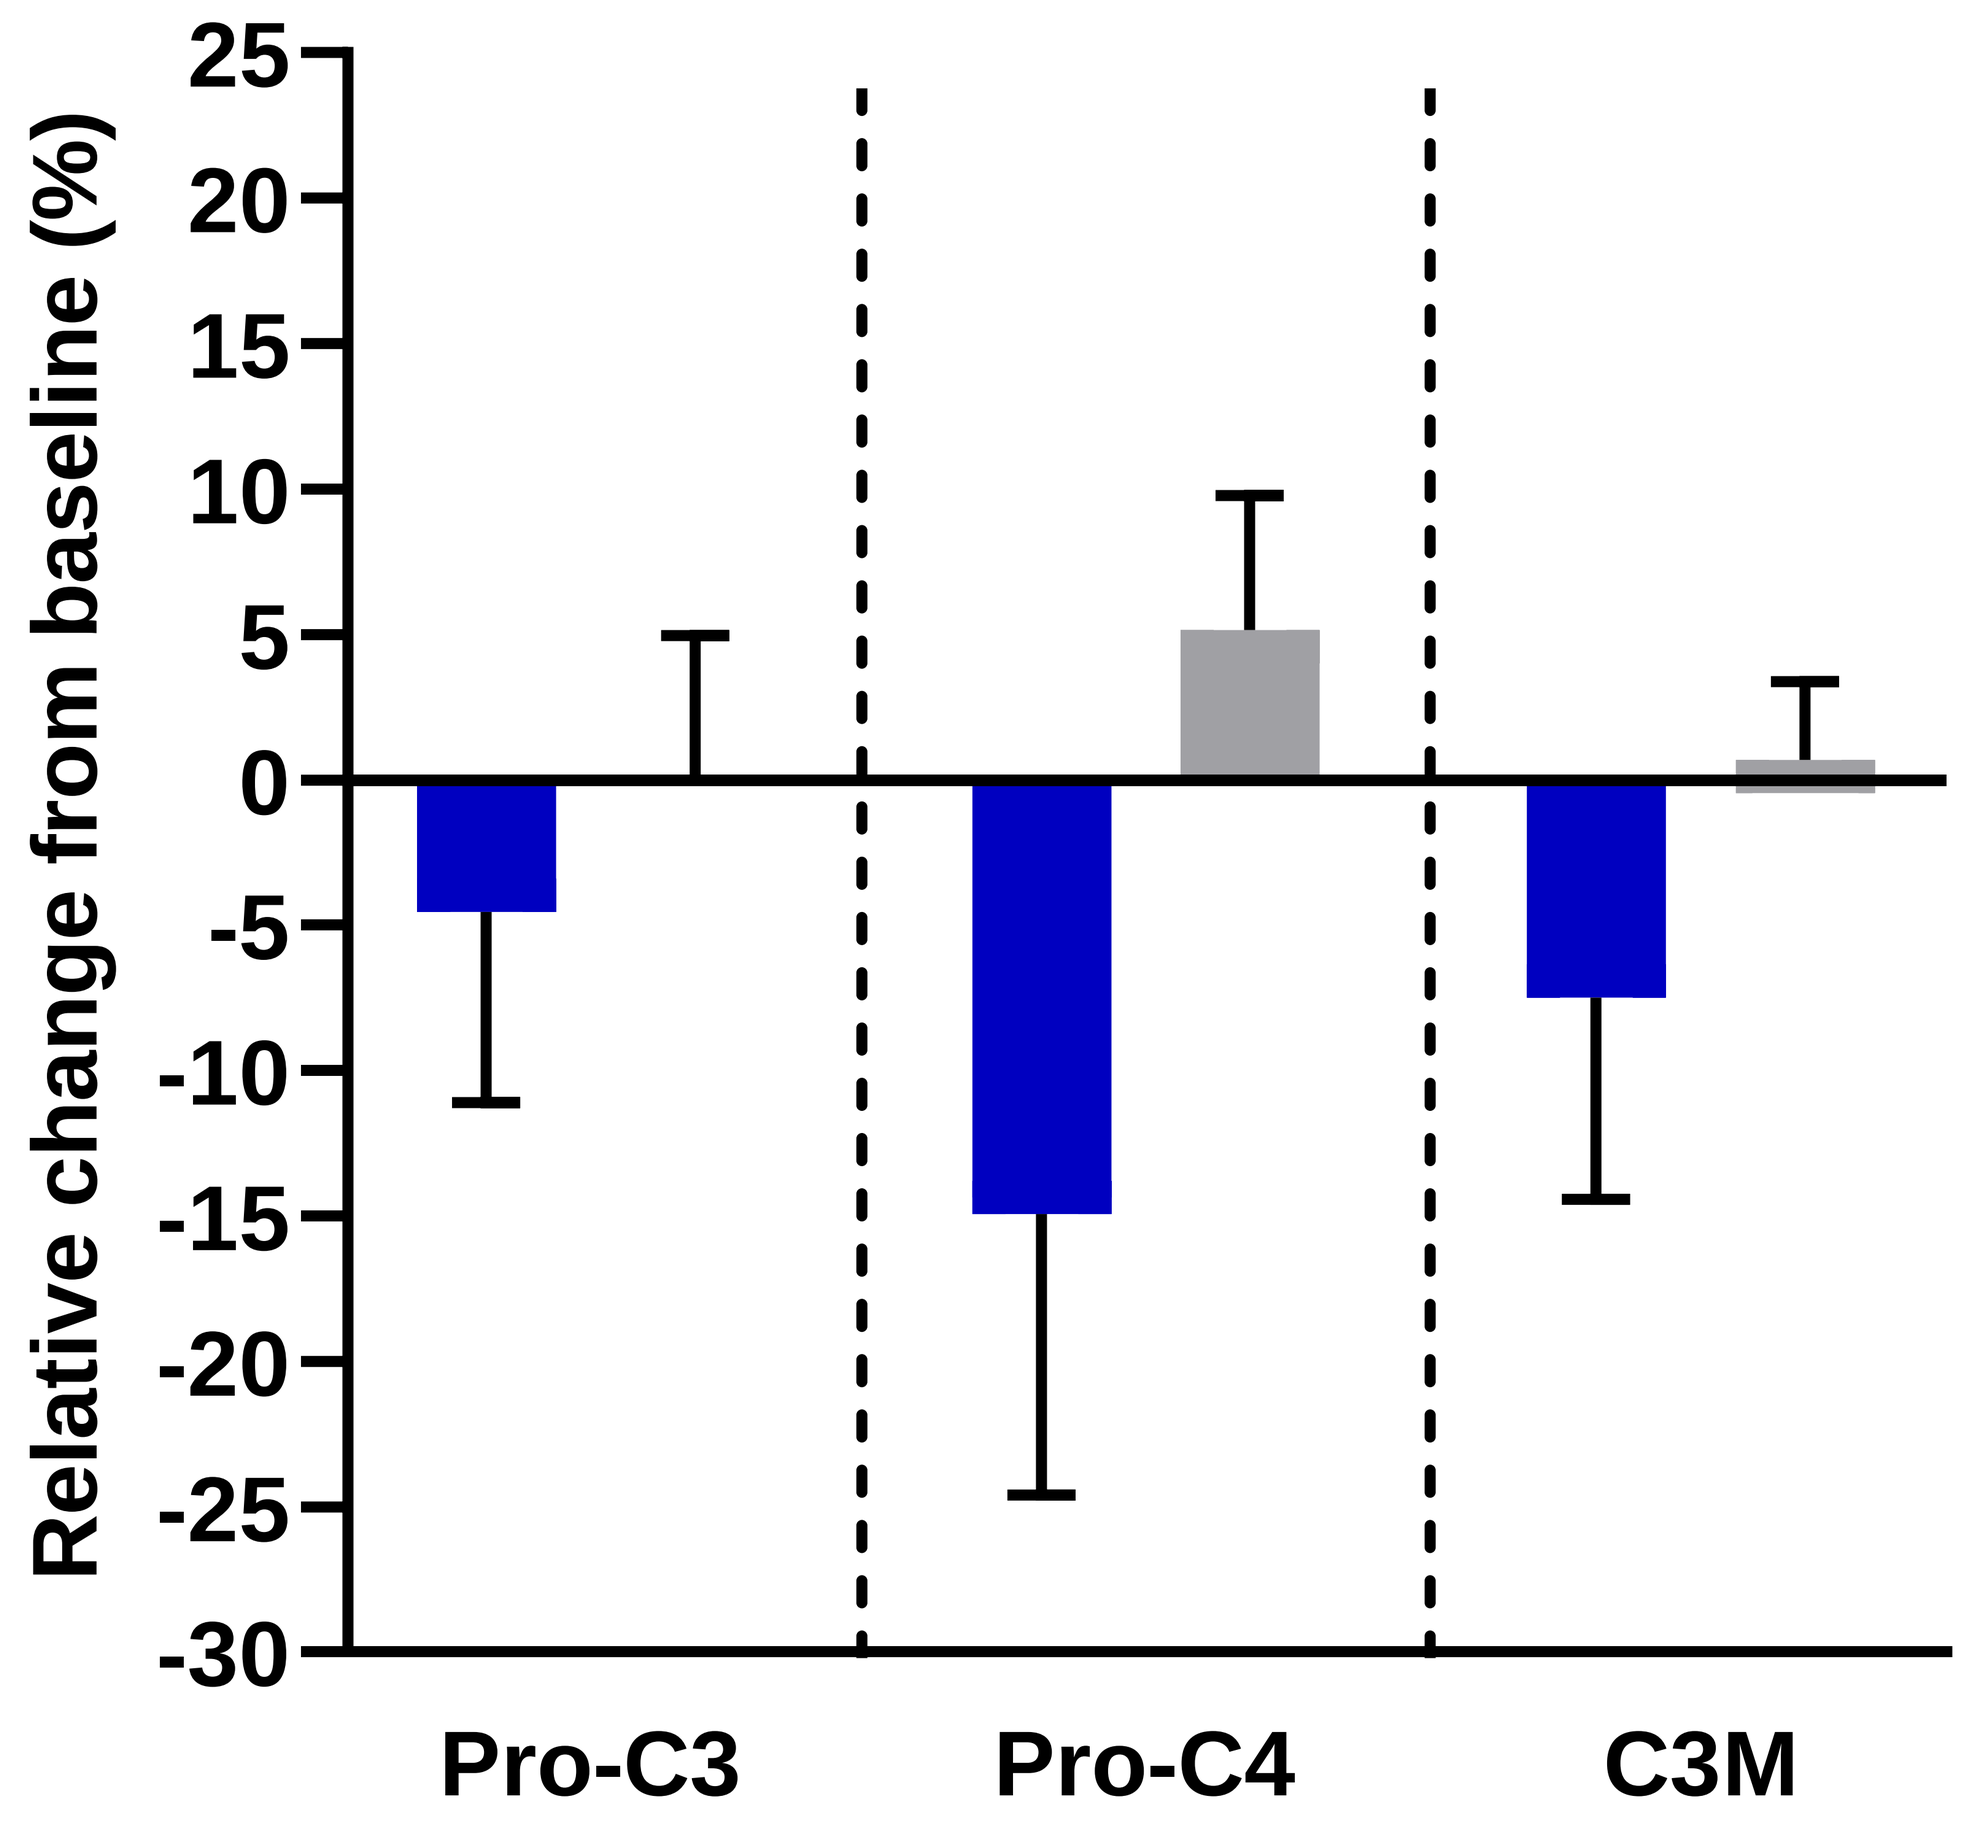
Relative change from baseline (%)
25
20
15
10
5
0
-5
-10
-15
-20
-25
-30
Pro-C3
Pro-C4
C3M

Serum fibrotic bio-markers*

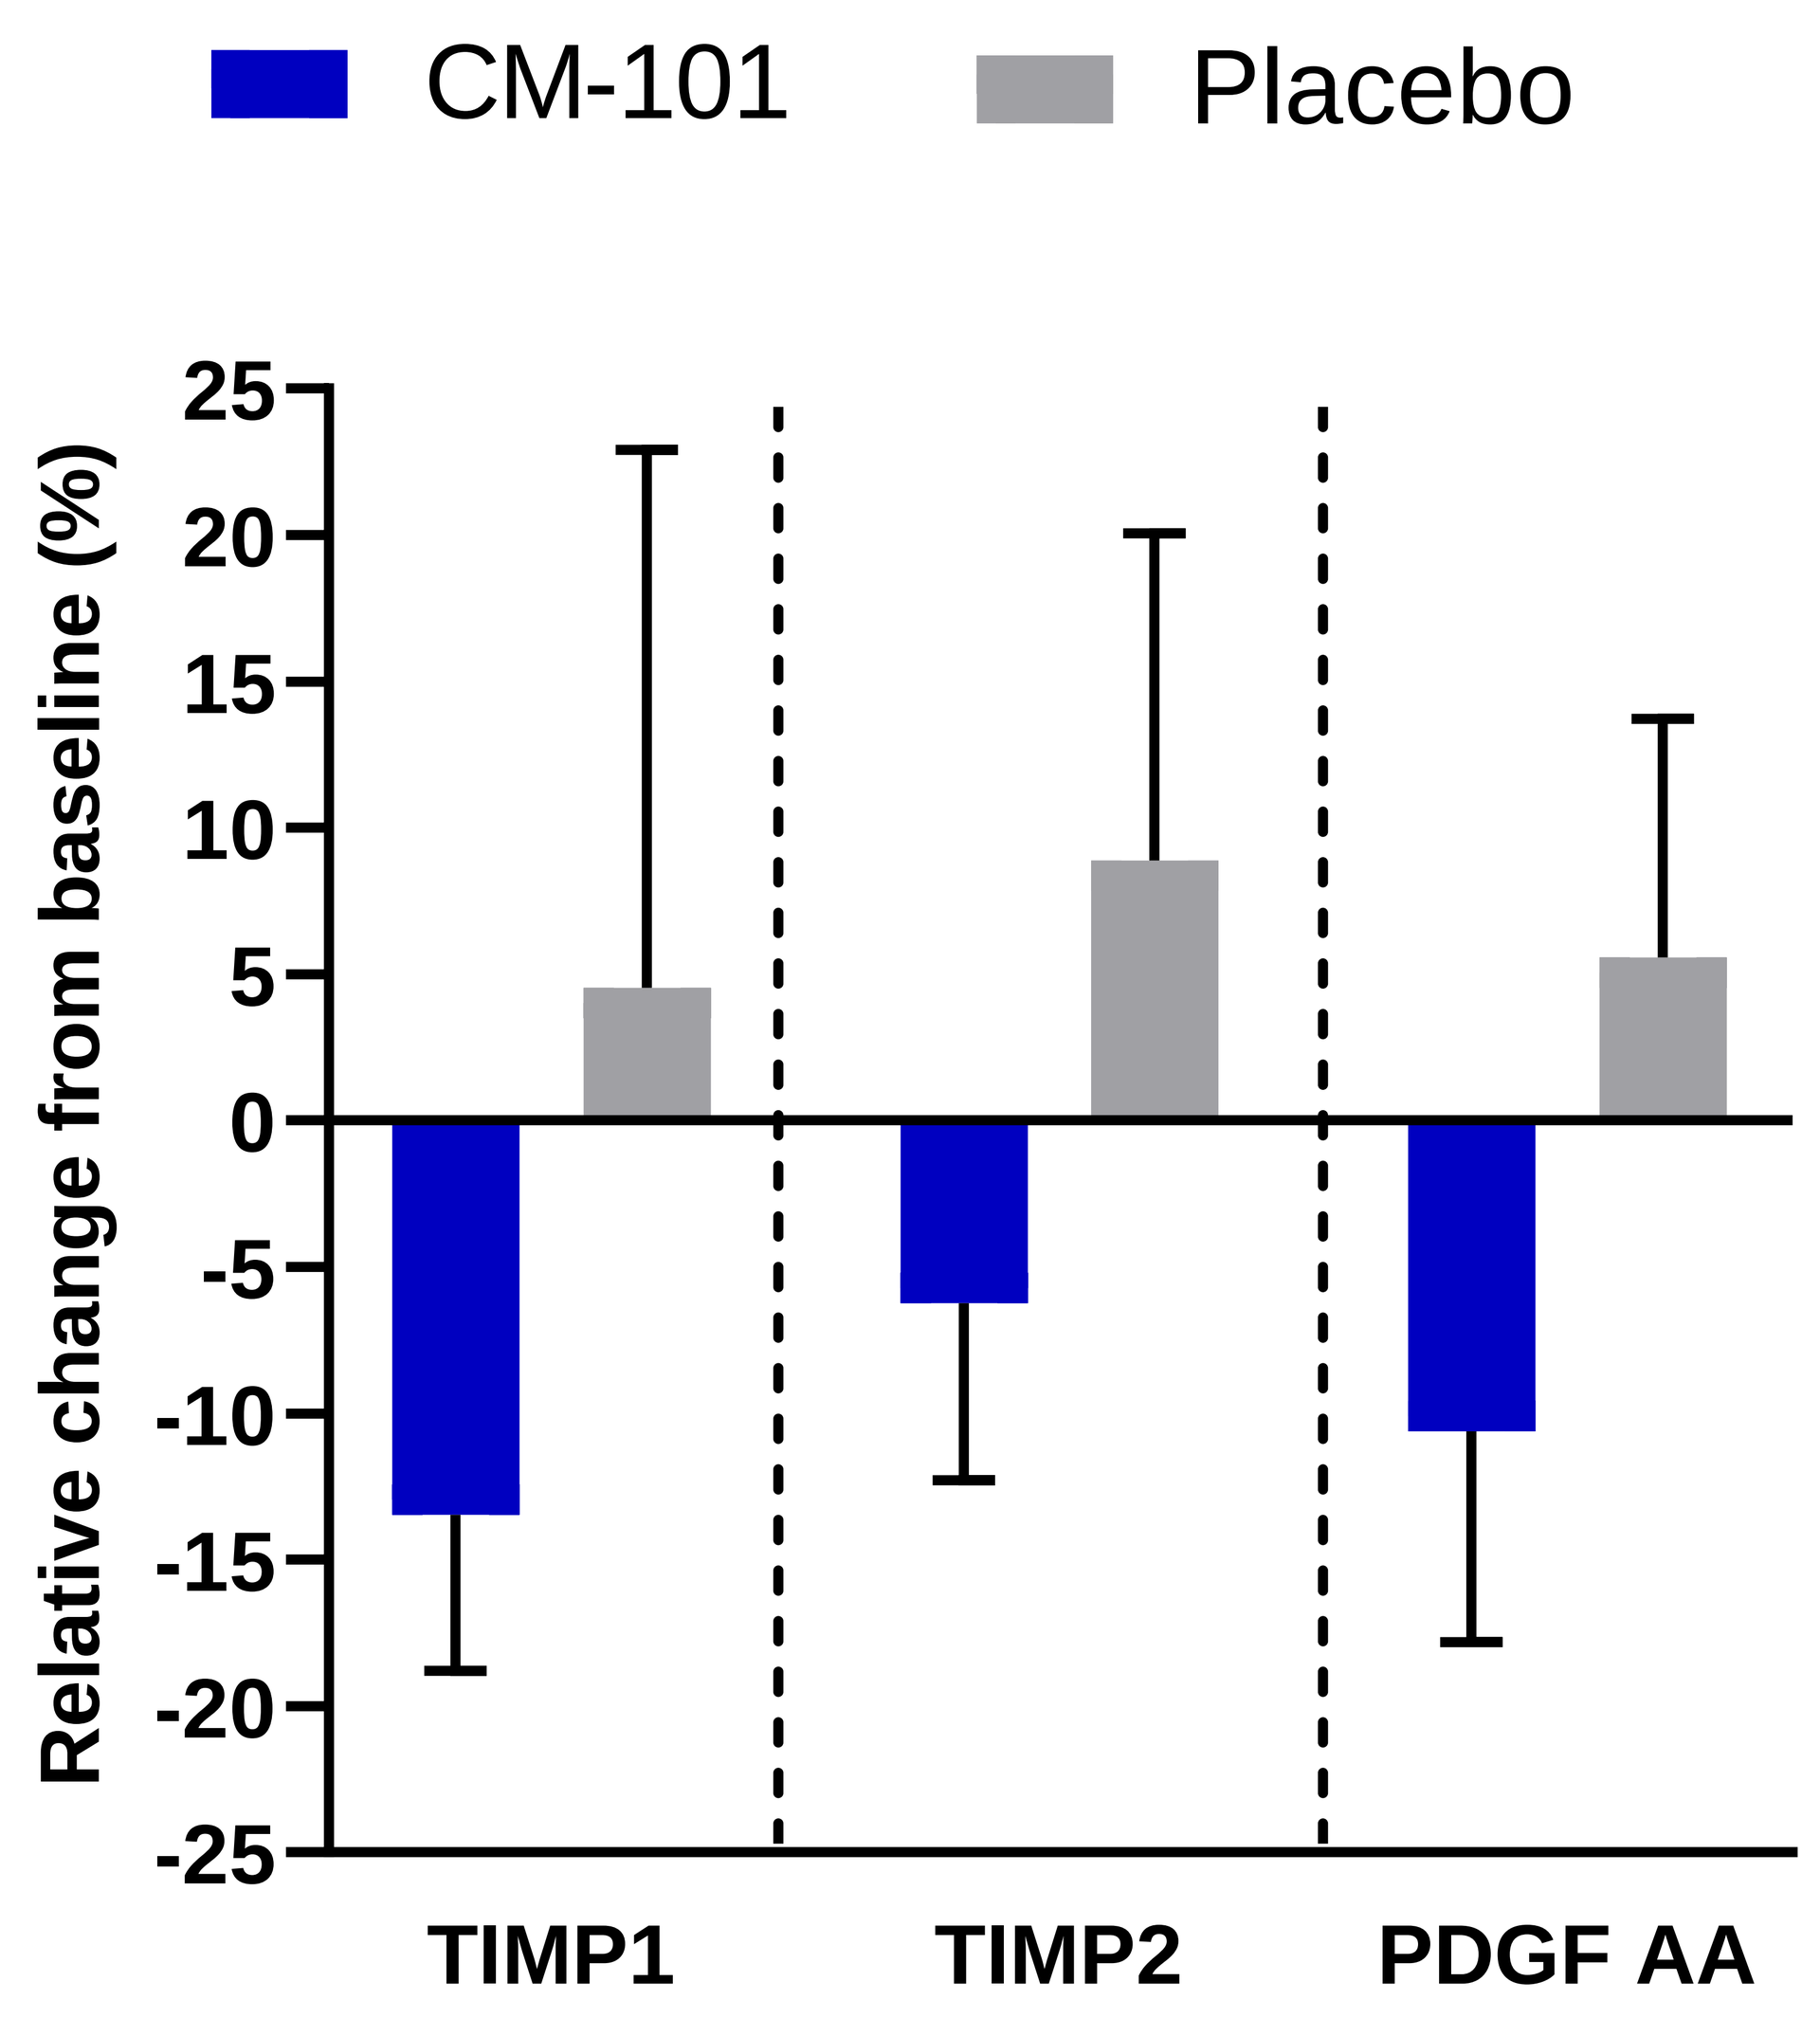
CM-101
Placebo
Relative change from baseline (%)
25
20
15
10
5
0
-5
-10
-15
-20
-25
TIMP1
TIMP2
PDGF AA

Elastography*

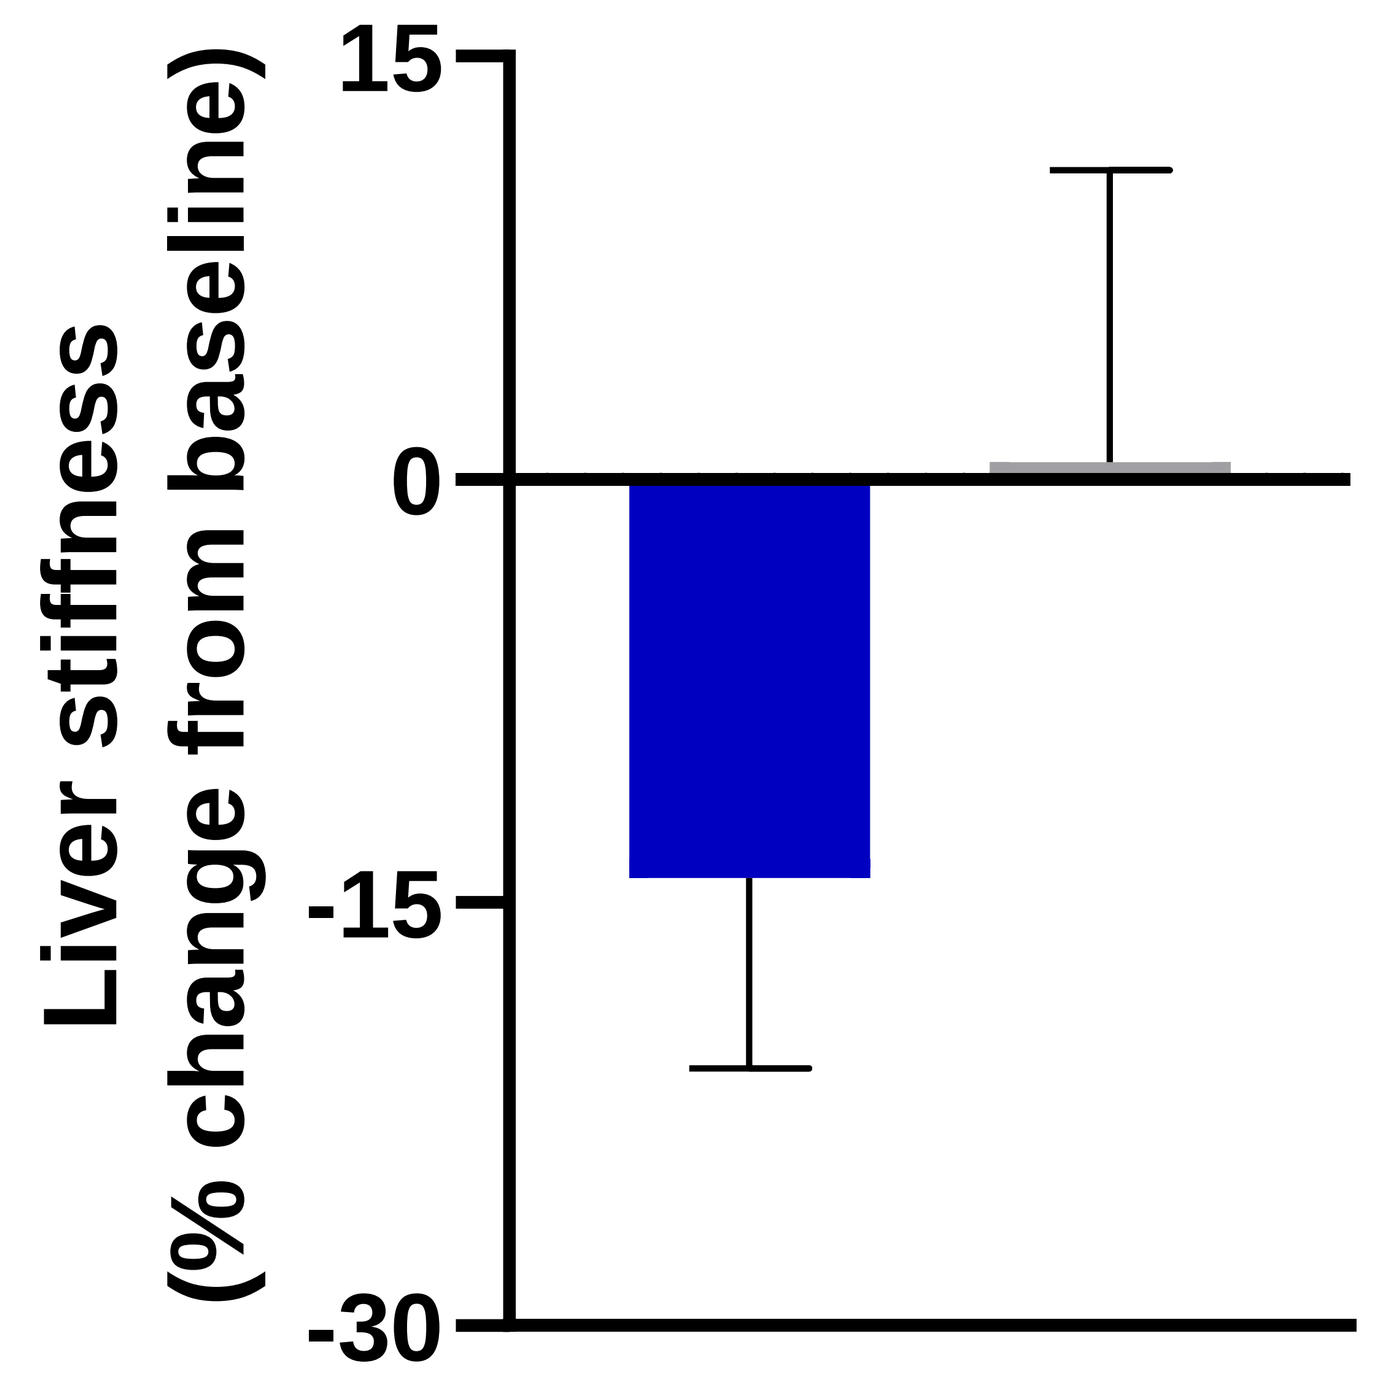
Liver stiffness
(% change from baseline)
15
0
-15
-30

PK

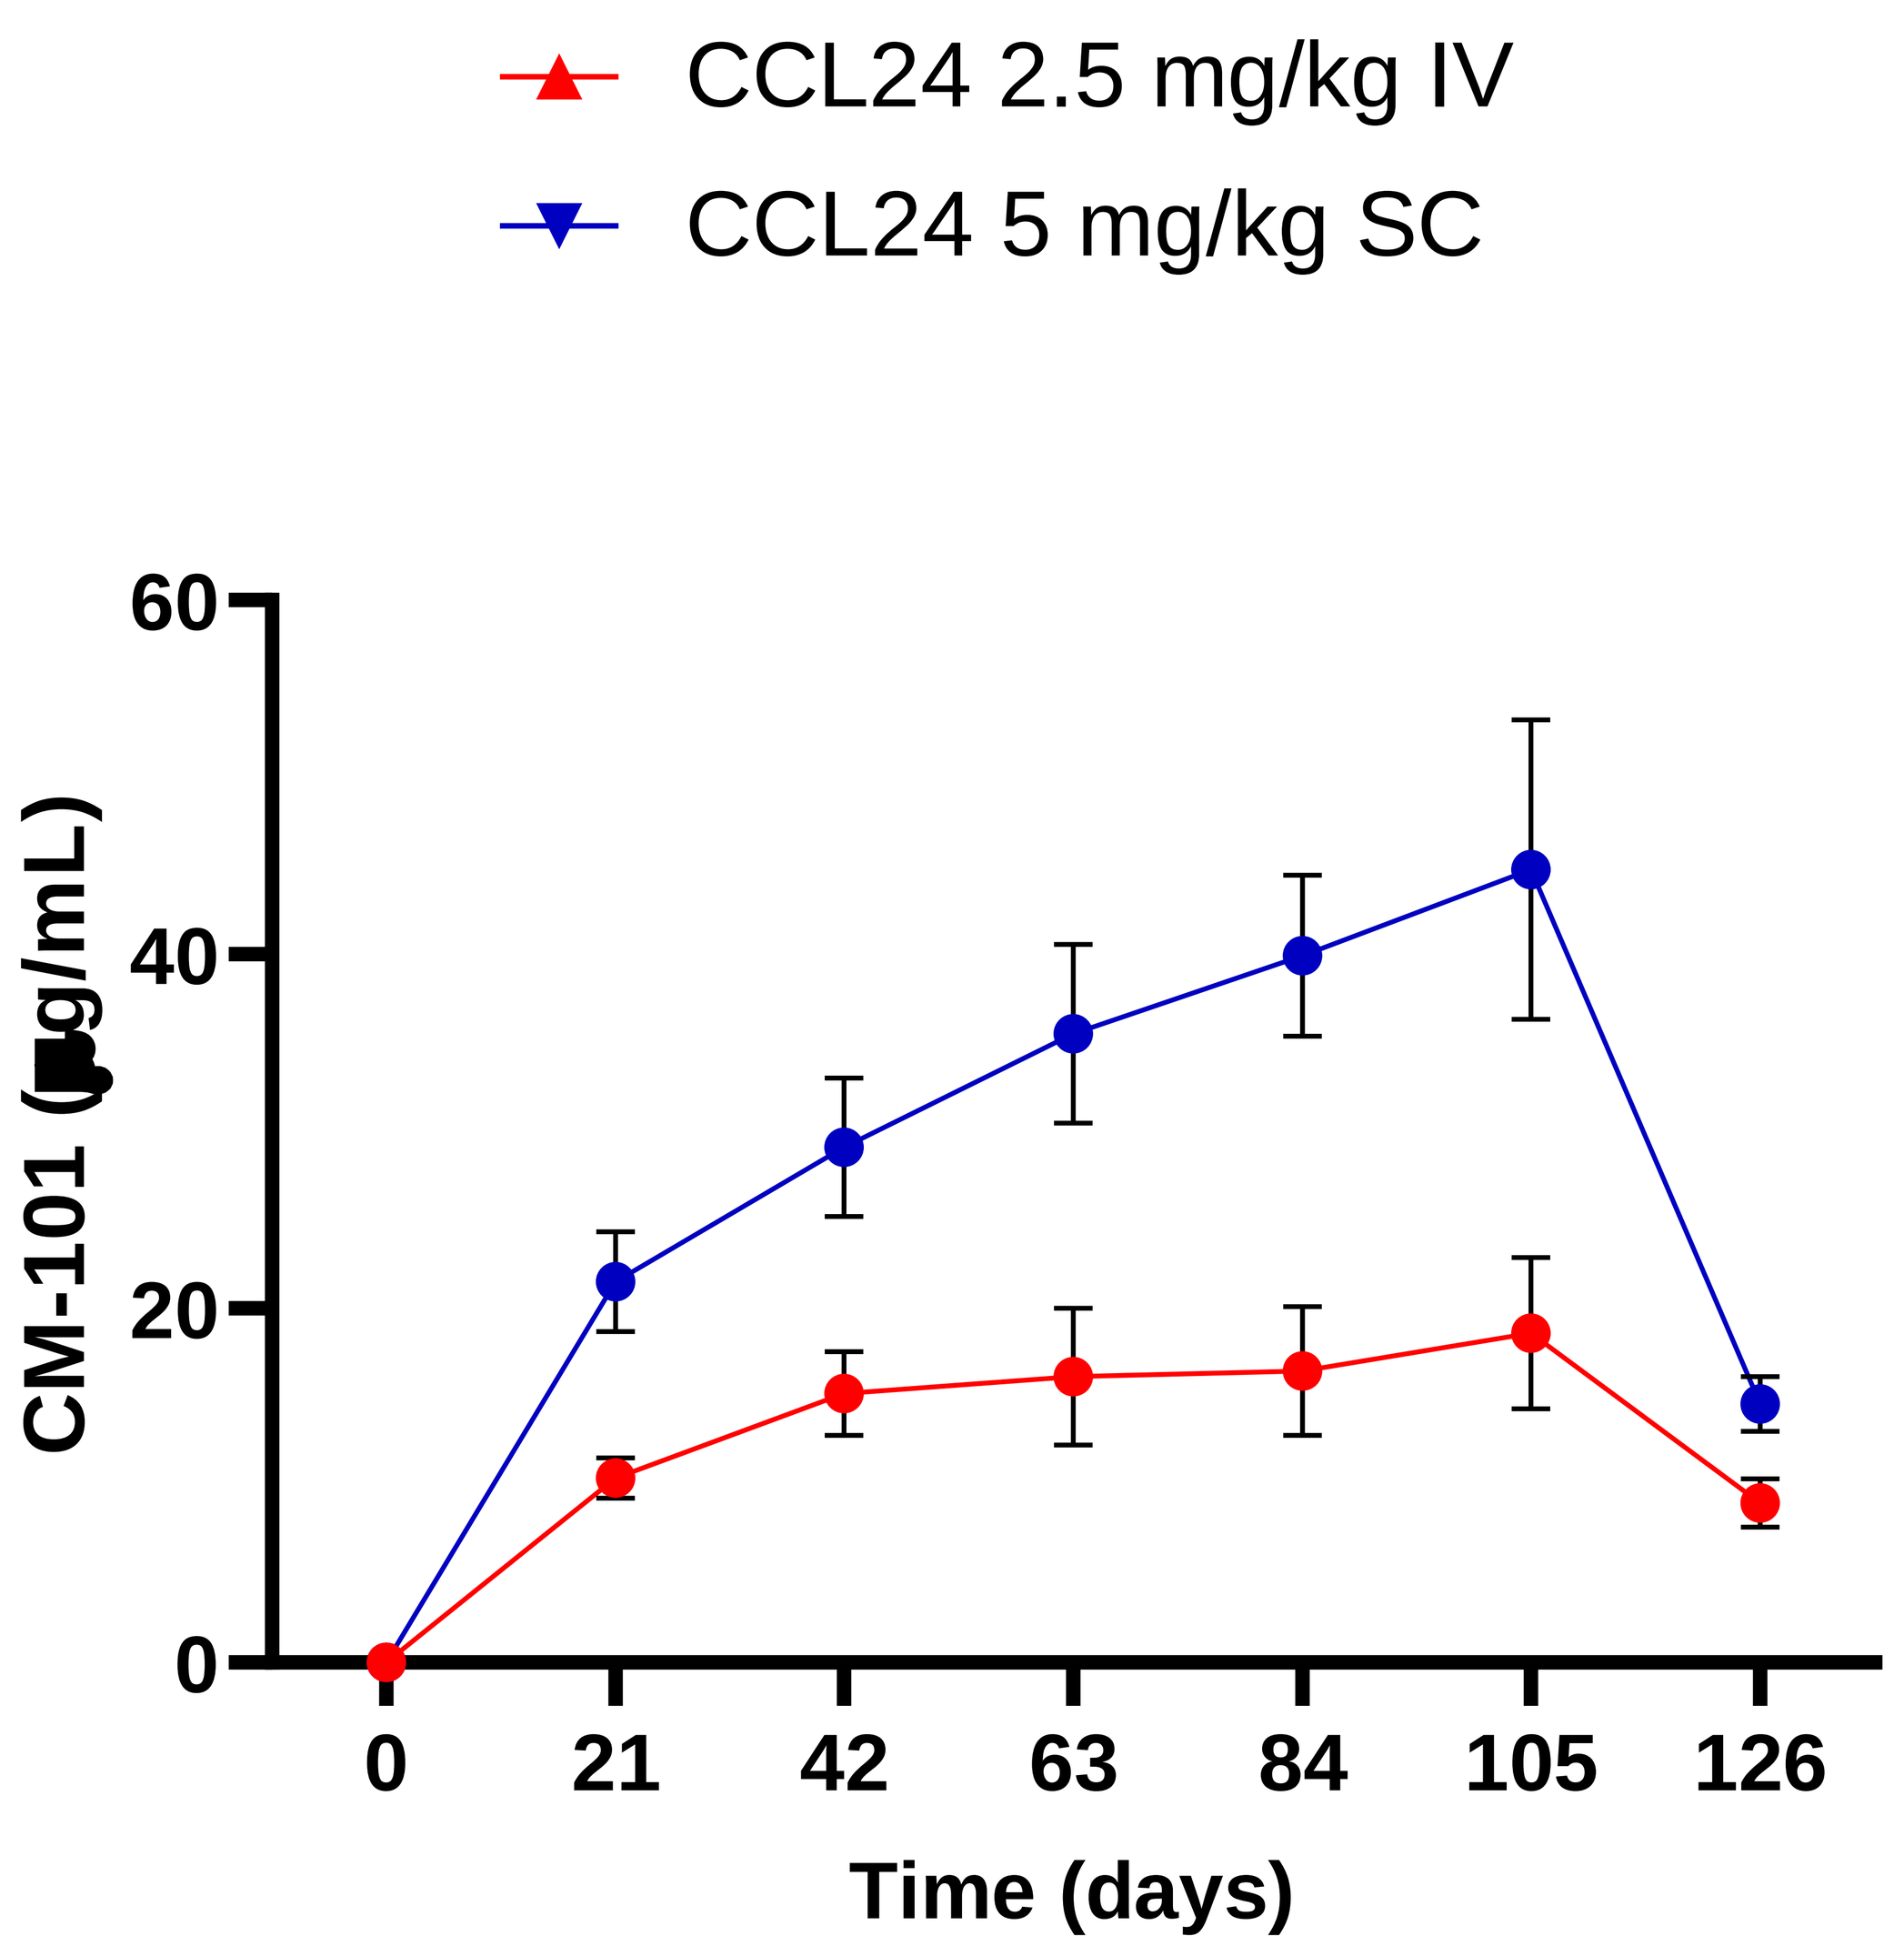
CCL24 2.5 mg/kg IV
CCL24 5 mg/kg SC
CM-101 (µg/mL)
60
40
20
0
0
21
42
63
84
105
126
Time (days)

Following treatment, a dose proportional increase in serum CM-101 levels was shown.

Following CM-101 treatment reduction in collagen turnover and fibrotic biomarkers was noted. For each marker change from baseline (%) is presented as median value ± IQR.

Pro-C3 is the N-terminal pro-peptide of Collagen type III (neo-peptide), Pro-C4 represent the internal epitope in 7S domain of type IV collagen, both represent fibrogenesis. C3M is a neo-epitope of MMP-9 mediated degradation of type III collagen that reflect the liver inflammatory state. Tissue inhibitor of metalloproteinases-1 and 2 (TIMP1, TIMP2) and the growth factor PDGF-AA are known fibrosis markers.

CM-101 treatment resulted in reduction of Pro-C3 (-4.5%), Pro-C4 (-11.8%) and C3M (-4.7%) compared to no change or slight elevation in the placebo treated group. Similarly, TIMP1, TIMP2 and PDGF AA were reduced by 10.5% , 4.4% and 10.6% respectively, while placebo group maintained a similar pattern of slight elevation. FibroScan™ data collaborates with the effect on bio-markers with a mean reduction of 14.1% in liver stiffness.

*Patients that had baseline elastography (FibroScan™) that was >4 kPA (n=10 CM-101; n=3 placebo).